OARSI 2017
ABS 680

PROTECTIVE ACTIONS OF MICROBIOTA INTERVENTION IN TWO ANIMAL MODELS OF OSTEOARTHRITIS: COMPLEMENTARY APPROACHES IN SPONTANEOUS OA IN DUNKIN HARTLEY GUINEA PIGS AND SURGICALLY INDUCED IN RABBIT

Y. Henrotin [1], S. Patrier [2], A. Pralus [2], M. Roche [2], A. Nivoliez [2]

[1]Bone and Cartilage Research Unit, Arthropôle Liege, University of Liege, Liege, BELGIUM [2]Département de Recherche et Développement, Biose®, Arpajon-sur-Cère, FRANCE

PURPOSE The aim of this study was to evaluate for the first time the potential protective effect of a lyophilized inactivated culture (LIC) or a lipoprotein extract (LIP) from Bifidobacterium longum CBi0703 which modulate microbiota regulation in both a spontaneous model of osteoarthritis (OA) in Guinea pigs and a surgically-induced OA model in rabbit. This kind of intervention with microbiota was shown to act on dysbiosis in chronic conditions. Histology of cartilage and synovial membrane and macroscopy of the articular structures were the primary endpoints. Soluble biomarkers were also considered to evaluate the treatment efficacy.

METHODS LIC and LIP (1 µg/kg/day) with or without vitamin C (1 mg/kg/day) were tested in Dunkin Hartley Guinea pigs (GP) spontaneously developing OA. Treatment were administered daily via oral route between 16 and 28 weeks. LIP (1-100 µg/kg/day) was further tested in a more aggressive model in rabbit after anterior cruciate ligament transection (ACLT) surgery of the right knee (7 weeks of treatment initiated 7 days after surgery, 8 weeks post-surgery follow up). It was administered daily in drinking water. All treatment groups were compared to placebo group. Macroscopy of the articular structures (rabbit only) and histology (cartilage and synovial membrane) were performed according to OARSI recommendations[a,b]. The kinetic of several biomarkers (Coll2-1, PIIANP, Fib3-2 and osteocalcin) was determined in Guinea pigs sera throughout the study.

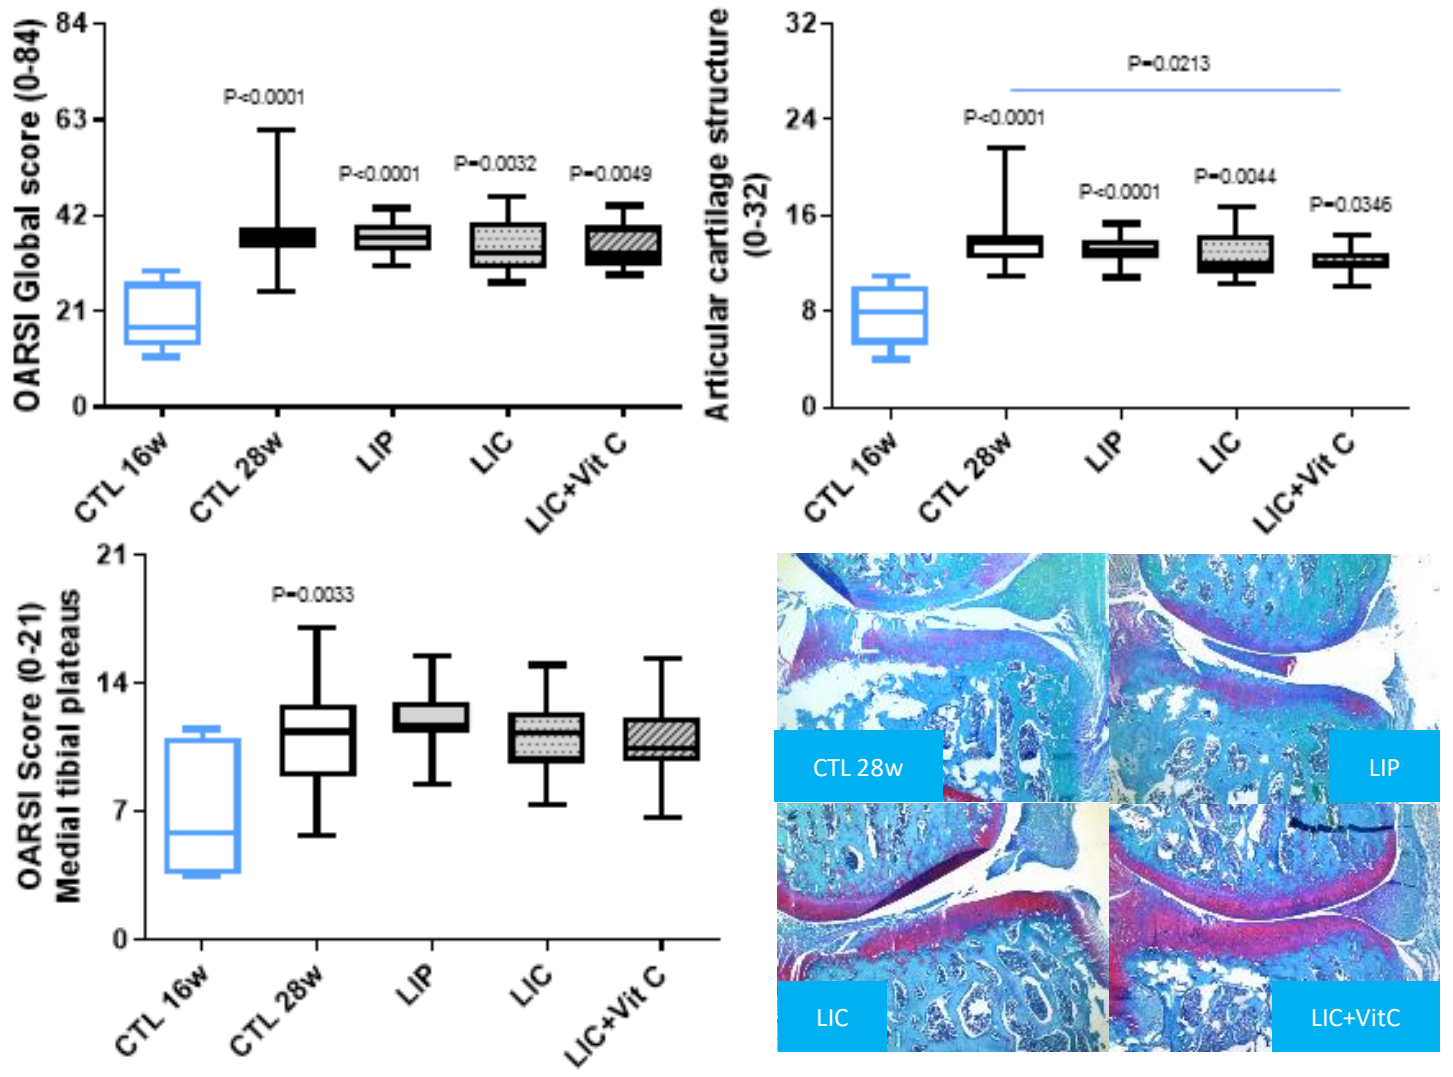

OARSI Global score (0-84)
P<0.0001
P<0.0001
P=0.0032
P=0.0049
Articular cartilage structure (0-32)
P=0.0213
P<0.0001
P<0.0001
P=0.0044
P=0.0346
OARSI Score (0-21) Medial tibial plateaus
P=0.0033
CTL 16w
CTL 28w
LIP
LIC
LIC+Vit C
CTL 28w
LIP
LIC
LIC+VitC

Figure 1. Histological score in right and left knees of GP. LIC and LIP tended to protect against the development of OA cartilage lesions. LIC plus vitamin C significantly maintain articular cartilage structure in comparison to placebo (p=0,0213 versus CTL 28w). The effect was even more pronounced when considering the medial tibial plateau alone. N=5-12, Kruskal-Wallis followed by Dunn's multiple comparison test versus CTL 16 weeks except otherwise specified.

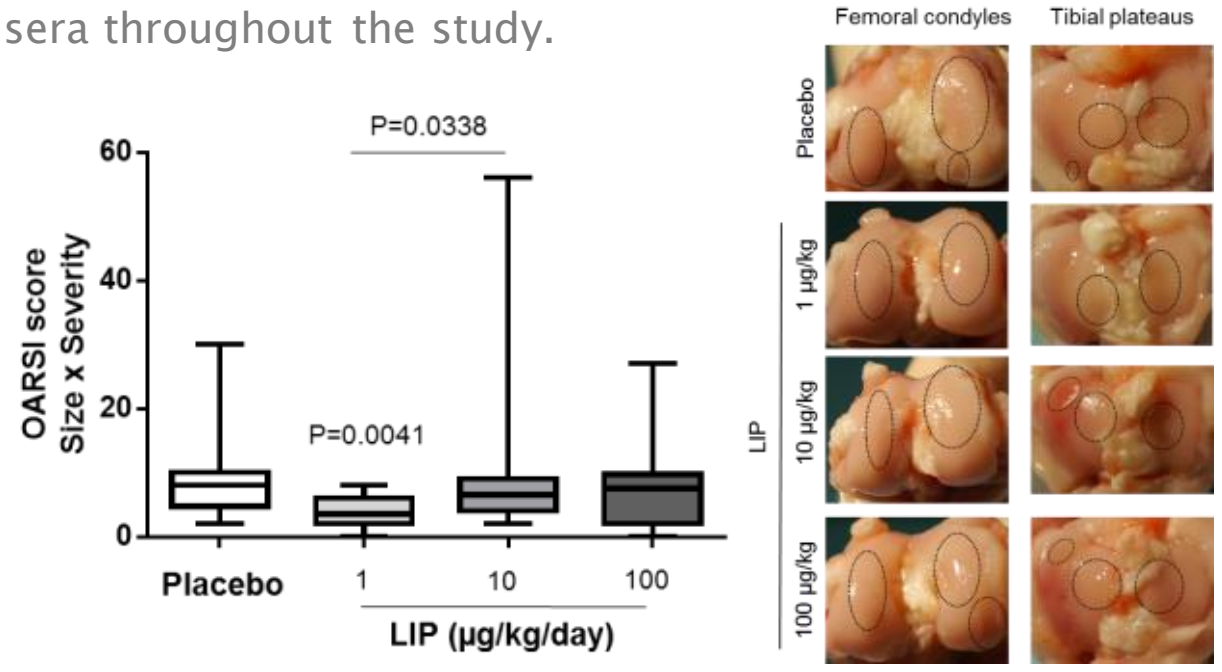

Femoral condyles
Tibial plateaus
P=0.0338
P=0.0041
OARSI score Size x Severity
Placebo
1
10
100
LIP (µg/kg/day)
Placebo
LIP
1 µg/kg
10 µg/kg
100 µg/kg

Figure 2. Macroscopic score in rabbit. The lowest tested dose (1 µg/kg) significantly reduced the macroscopic score (p-0.041). N=10, Kruskal-Wallis followed by Dunn's multiple comparison test versus placebo except otherwise specified.

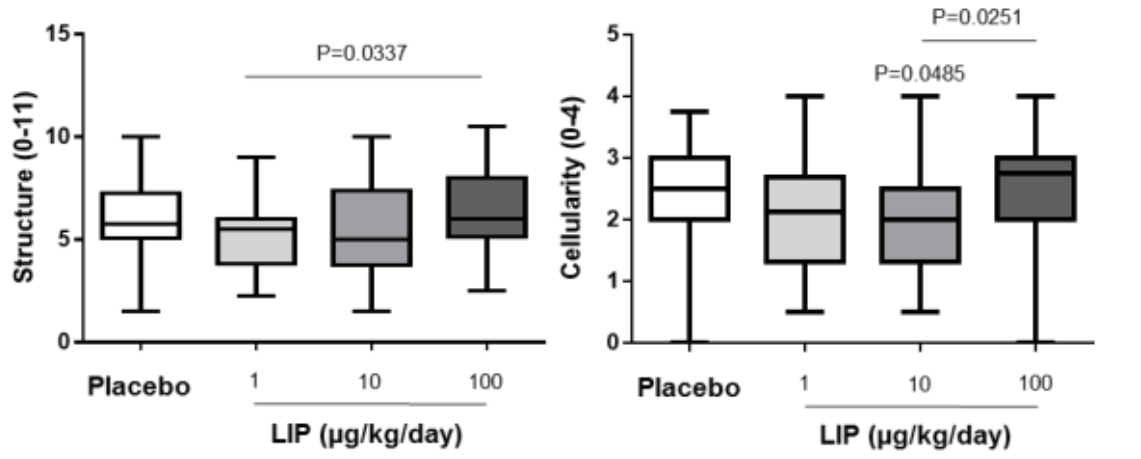

Structure (0-11)
P=0.0337
Cellularity (0-4)
P=0.0251
P=0.0485
Placebo
1
10
100
LIP (µg/kg/day)

Figure 3. Histological score in rabbit. The lowest tested doses (1-10 µg/kg) reduced both the structure and cellularity scores. N=10, Kruskal-Wallis followed by Dunn's multiple comparison test versus placebo except otherwise specified.

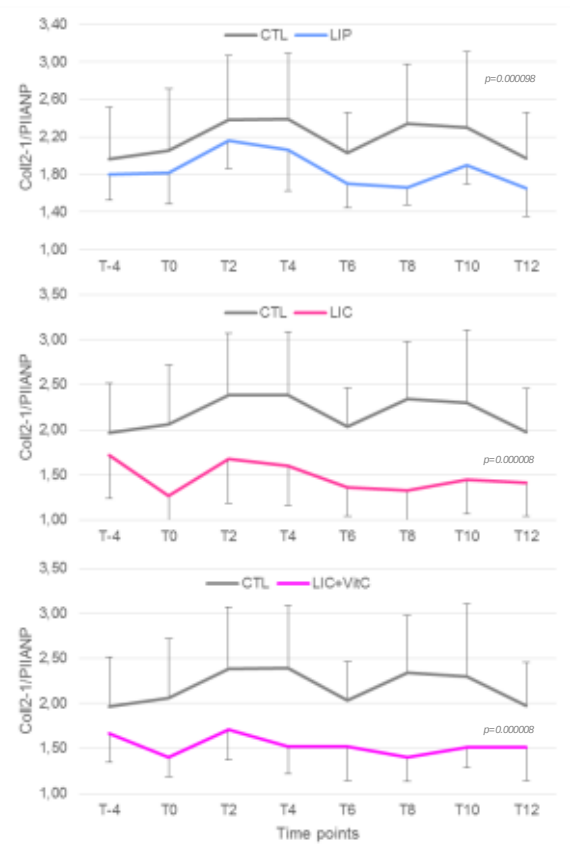

CTL
LIP
p=0.000098
LIC
p=0.000008
LIC+VitC
p=0.000008
Coll2-1/PIIANP
Time points

Figure 4. Coll2-1/PIIANP: Coll2-1, marker of type II collagen degradation and PIIANP, marker of type II collagen synthesis. Coll2-1 was reduced whereas PIIANP was increased by LIP and LIC (±VitC). Coll2-1/PIIANP ratio was in favor of type II collagen synthesis. (ANOVA, n=10)

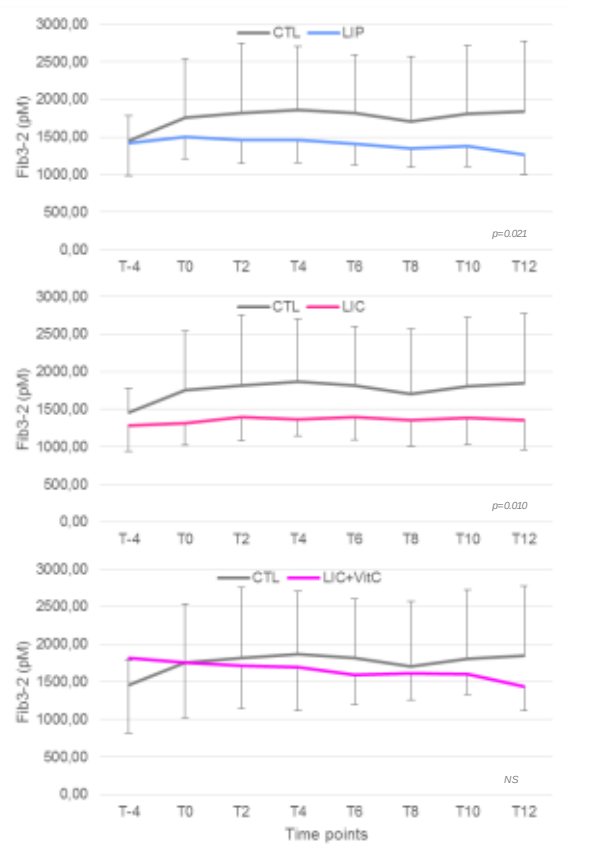

CTL
LIP
p=0.021
LIC
p=0.010
LIC+VitC
NS
Fib3-2 (pM)
Time points

Figure 5. Fib3-2, marker of cartilage degradation. LIP and LIC tended to decrease Fib3-2, another effect toward cartilage preservation. (ANOVA, n=10)

[a] Kraus et al, Osteoarthritis and Cartilage, 2010, 18:S35-S52

[b] Laverty et al, Osteoarthritis and Cartilage, 2010, 18:S53-S65

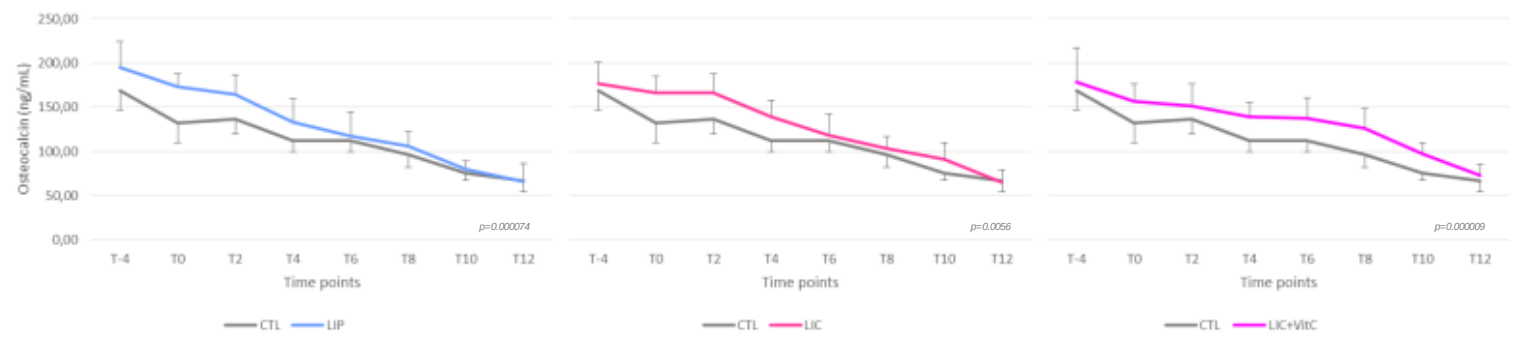

Osteocalcin (ng/mL)
p=0.000074
p=0.0056
p=0.000009
Time points
CTL
LIP
LIC
LIC+VitC

Figure 6. Osteocalcin, marker of bone turnover. LIP and LIC (±VitC) inhibited the reduction in osteocalcin levels observed in the placebo group. LIP and LIC (±VitC) enhanced bone turnover. (ANOVA, n=10)

CONCLUSIONS This study produced interesting and promising results by the dual approach in two complementary models of OA. The results provided by biomarkers assays were interesting and valuable for the interpretation of the study results in addition to the gold standard approach recommended by OARSI. LIC could be considered as a preventive treatment such as dietary supplement to protect against OA development and can be foreseen as a new non-pharmacological treatment against OA with or without the addition of vitamin C. This study is the first one to demonstrate a potential effect of microbiota acting most probably on dysbiosis during the disease pathogenesis and open a new therapeutic approach.