Interleukin-1, interleukin-6 and oncostatin M stimulate normal subchondral osteoblasts to induce cartilage degradation.

Christelle Sanchez[1], Michelle A. Deberg[1], Nathalie Piccardi[2], Philippe Msika[2], Jean-Yves L. Reginster[1] and Yves E. Henrotin[1]

[1] Bone and Cartilage Research Unit, Liège, Belgium

[2] Laboratoires Expanscience, Epernon, France

Introduction. Previously, we have demonstrated that osteoblasts from the sclerotic subchondral bone are phenotypically different that those from the non sclerotic area. Sclerotic osteoblasts produced more IL-6, TGF-β1, ALP and OC but similar amount of IL-1β than non sclerotic osteoblasts.

Aim of Study. To determine the effects of osteoarthritic subchondral osteoblasts on the metabolism of human chondrocytes in alginate beads.

Methods. Human chondrocytes were isolated from OA cartilage and cultured in alginate beads for 4 days in the absence or in the presence of OA subchondral osteoblasts in monolayer (co-culture system, figure 1). Before co-culture, osteoblasts were cultured for 72 hours with or without 1.7 ng/ml interleukin (IL)-1β, 100 ng/ml IL-6 with its soluble receptor (IL-6sR; 50 ng/ml) or 10 ng/ml oncostatin M (OSM). Aggrecan and matrix metalloproteases (MMP) -3 and -13 mRNA levels in chondrocytes were quantified by real time polymerase chain reaction. Aggrecan production was assayed by a specific enzyme amplified sensitivity immunoassay (EASIA).

Co-culture system

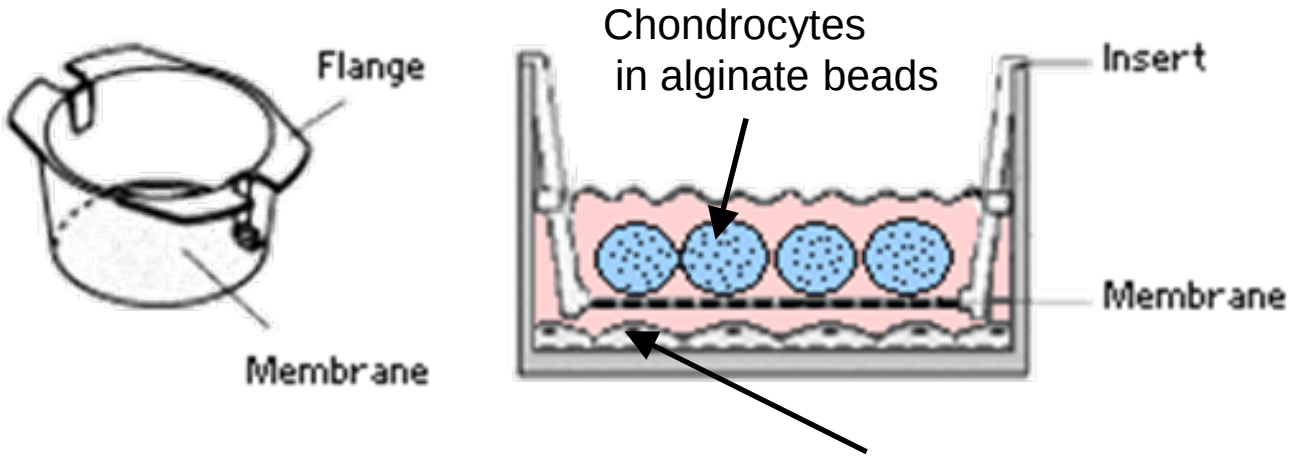
Flange
Membrane
Chondrocytes
in alginate beads
Insert
Membrane
Subchondral osteoblasts

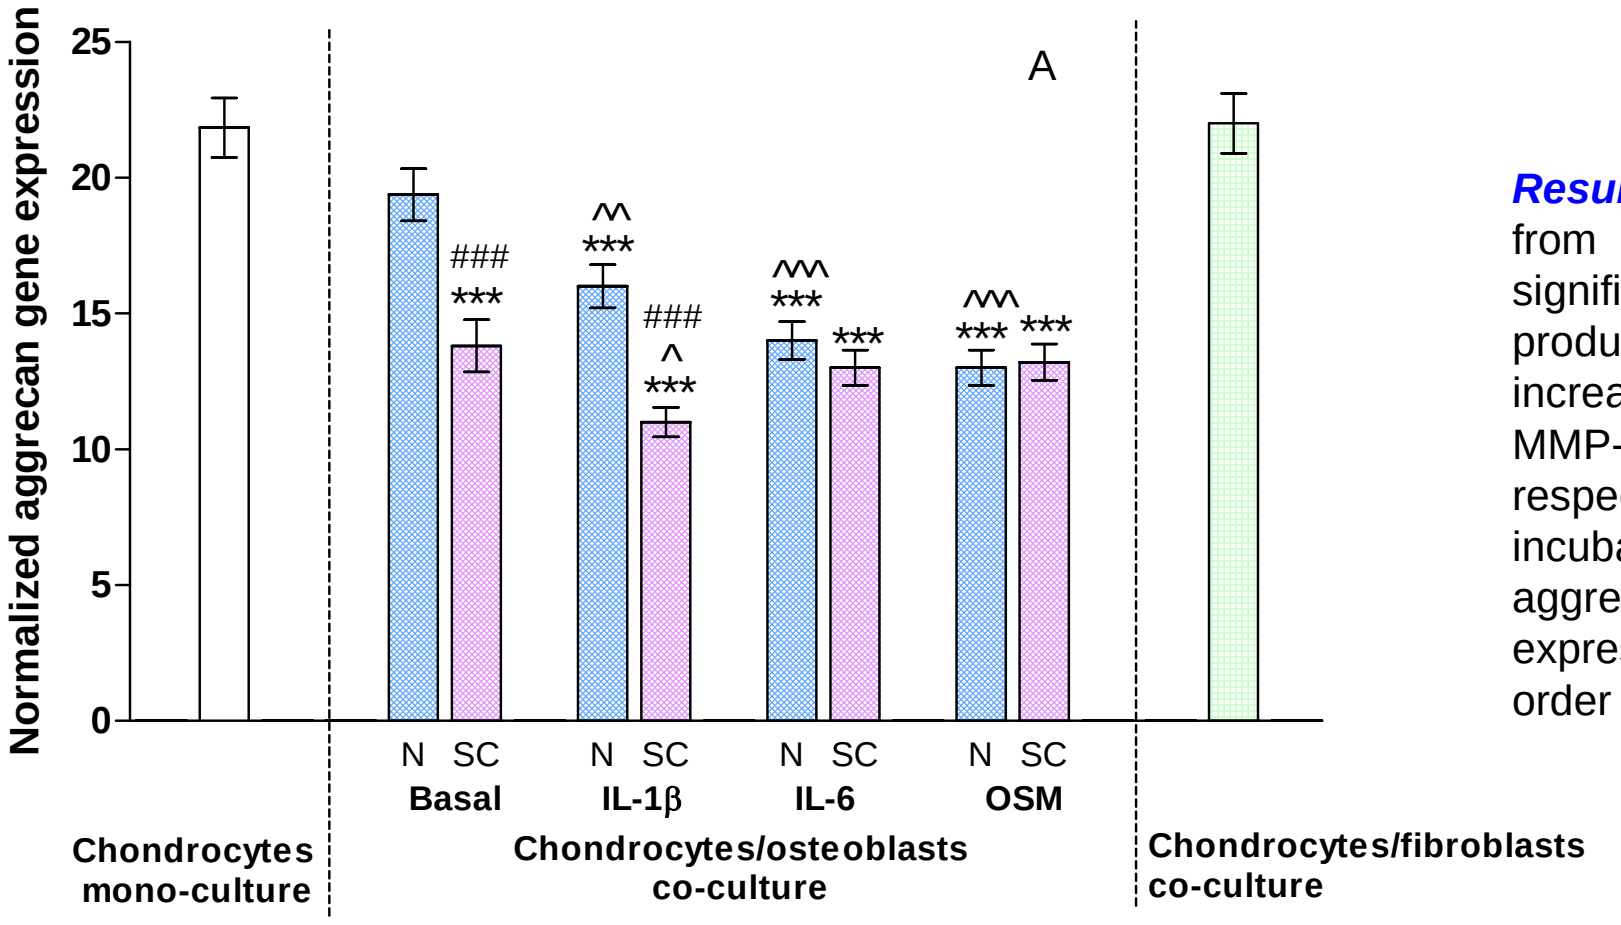
Normalized aggrecan gene expression
A
0
5
10
15
20
25
N SC
Basal
IL-1β
IL-6
OSM
Chondrocytes mono-culture
Chondrocytes/osteoblasts co-culture
Chondrocytes/fibroblasts co-culture

Results. Osteoblasts isolated from sclerotic (SC) but not from non sclerotic (N) zones of subchondral bone significantly decreased (-28 %, p<0.001) aggrecan production and aggrecan gene expression and significantly increased by human OA chondrocytes in alginate beads MMP-3 and MMP-13 gene expression (1.65 and 2 times, respectively, p < 0.001) (figure 2). When they were pre-incubated with IL-1β, IL-6 or OSM, N osteoblasts inhibited aggrecan synthesis and increased MMP-3 and -13 gene expression by chondrocytes in alginate beads in a same order of magnitude that SC osteoblasts.

*Figure 1: GADPH-normalized AGG (A), MMP-3 (B) and MMP-13 (C) gene expression by chondrocytes cultured 4 days with or without osteoblasts from non sclerotic (N) or sclerotic (SC) subchondral bone zones or normal skin fibroblasts. Osteoblasts were pre-treated or not with 1.7 ng/ml IL-1β, 100 ng/ml IL-6 + 50 ng/ml IL-6sR or 10 ng/ml OSM during 72h before co-culture. Results are means of 3 co-culture experiments each realised in quadruplicate +/- SEM. * p<0.05, ** p<0.01 and *** p<0.001 between co-culture and mono-culture experiments, ## p<0.01 and ### p<0.001 between N and SC osteoblastes, ^ p<0.05, ^^ p<0.01 and ^^^ p<0.001 between basal and cytokine pre-treated osteoblasts.*

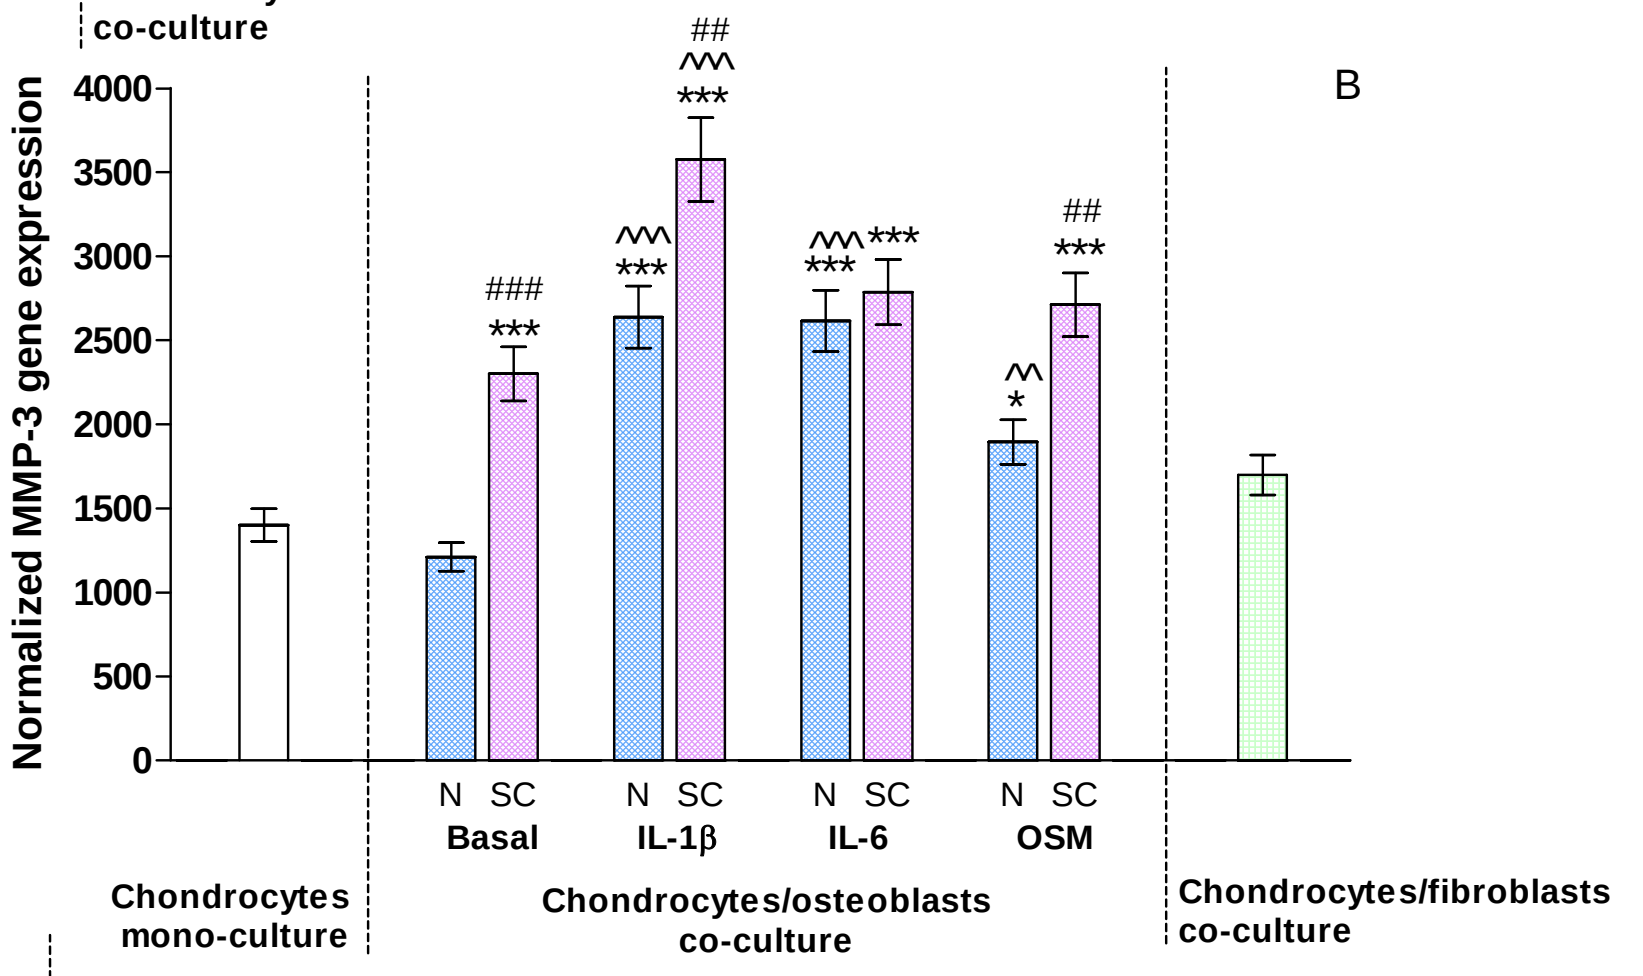
Normalized MMP-3 gene expression
B
0
500
1000
1500
2000
2500
3000
3500
4000
N SC
Basal
IL-1β
IL-6
OSM
Chondrocytes mono-culture
Chondrocytes/osteoblasts co-culture
Chondrocytes/fibroblasts co-culture

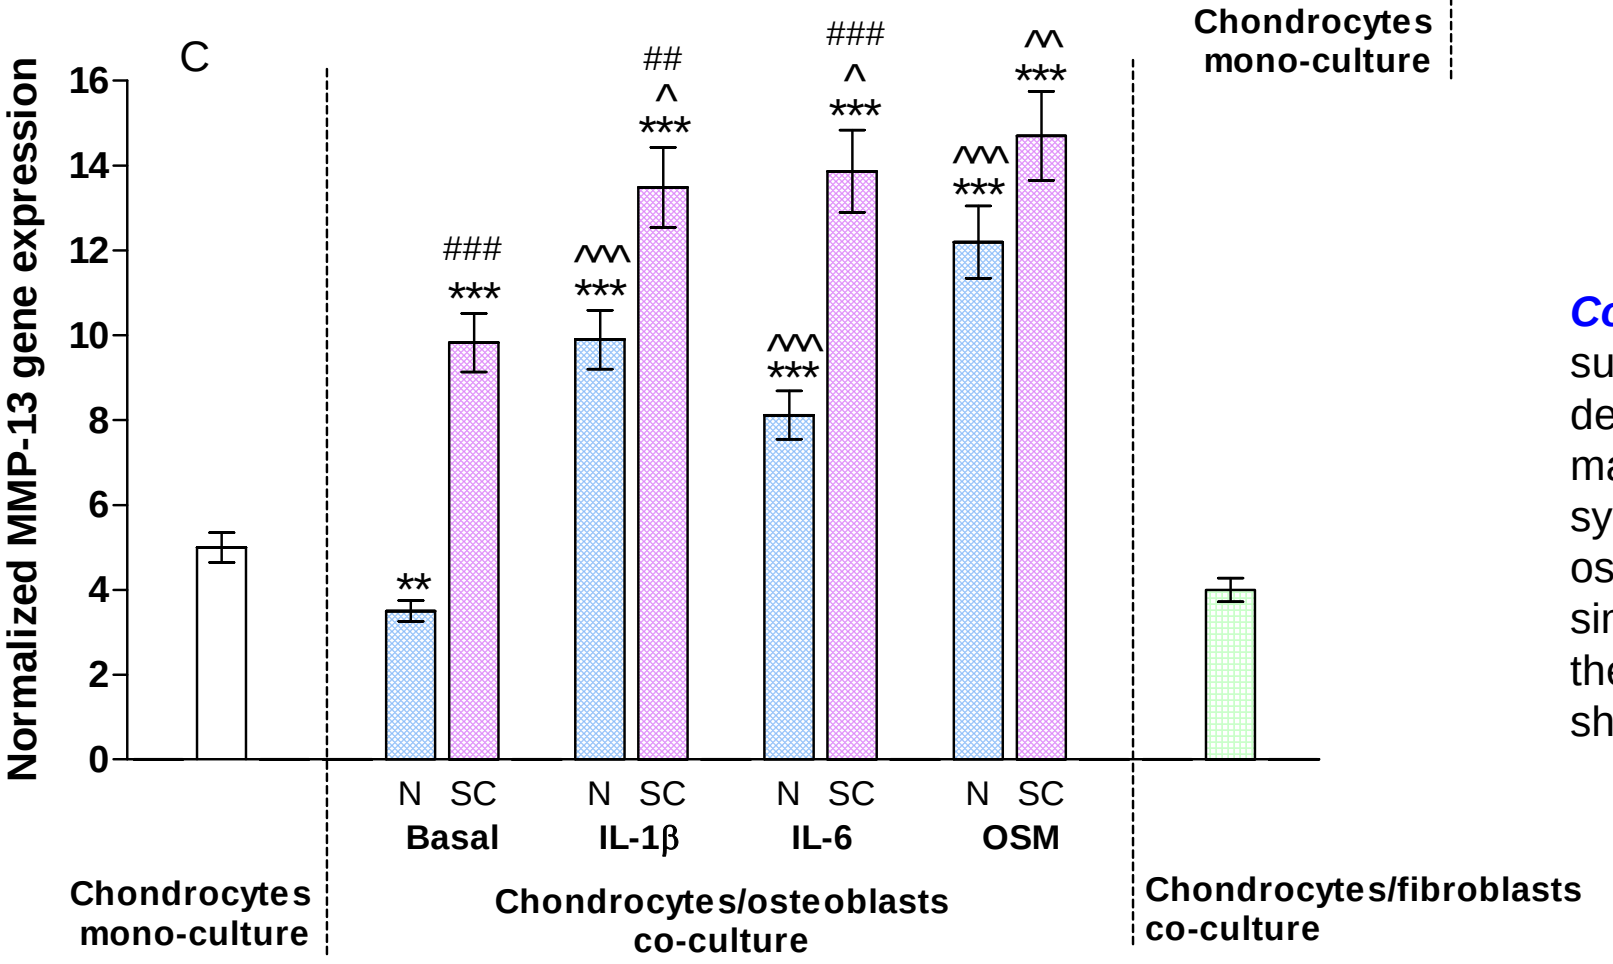
Normalized MMP-13 gene expression
C
0
2
4
6
8
10
12
14
16
N SC
Basal
IL-1β
IL-6
OSM
Chondrocytes mono-culture
Chondrocytes/osteoblasts co-culture
Chondrocytes/fibroblasts co-culture

Conclusions. These results demonstrate that OA subchondral osteoblasts could contribute to cartilage degradation by stimulating chondrocytes to produce more matrix metalloproteases and by inhibiting aggrecan synthesis. IL-1β, IL-6 and OSM may stimulate normal osteoblasts to induce chondrocyte metabolic dysregulation similar to those observed in OA cartilage, suggesting that these cytokines play an important role in the phenotype shift of subchondral osteoblasts in OA.