COMPRESSION INDUCES THE EXPRESSION OF A SCLEROTIC PHENOTYPE IN HUMAN SUBCHONDRAL OSTEOBLASTS

Christelle Sanchez[1], Odile Gabay[2], Laurence Pesesse[1], Philippe Msika[3], Caroline Baudoin[3], Yves Henrotin[1]

[1]Bone and Cartilage Research Unit, University of Liège, Belgium
[2]Cartilage Biology and Orthopaedic Branch NIH Bethesda, Washington, USA
[3]Laboratoires Expanscience, Epernon, France

Purpose. Recent data showed that subchondral bone plays an important role in osteoarthritis (OA). Metabolic and morphologic modifications in this tissue contribute to the degradation of the overlaying cartilage. It was suggested that abnormal mechanical pressure applied on the joint was responsible for these changes. Here, we evaluated the effects of compression on osteoblasts from subchondral bone.

Methods. Osteoblasts were isolated from sclerotic (SC) or non-sclerotic (NSC) areas of human OA subchondral bone. After 28 days, osteoblasts were surrounded by a newly synthesized matrix and formed a strong membrane. This osteoblasts-containing membrane was then placed onto a Biopress Flexercell plate and submitted to compression (1.67 MPa) for 4 hours at the frequency of 1 Hz. The expression of IL-6, IL-8, COX-2, VEGF, IGF-1, OPG and RANKL was evaluated by RT-PCR. IL-6, IL-8 and PGE_2 were quantified by ELISA.

Results. Basal IL-6, VEGF, COX-2, IGF-1 and RANKL mRNA levels were significantly increased in SC osteoblasts as compared to NSC. By contrast, SC osteoblasts expressed less OPG than those from NSC areas. Compressions induced the expression of genes coding for IL-6, IL-8, COX-2, IGF-1, VEGF and RANKL but decreased the expression of OPG in NSC osteoblasts ($p<0.01$). IL-6, IL-8 and PGE_2 productions were also stimulated by compressions. Interestingly, compressed NSC osteoblasts expressed similar levels of these genes than SC osteoblasts, suggesting that mechanical strains could be responsible for SC phenotype.

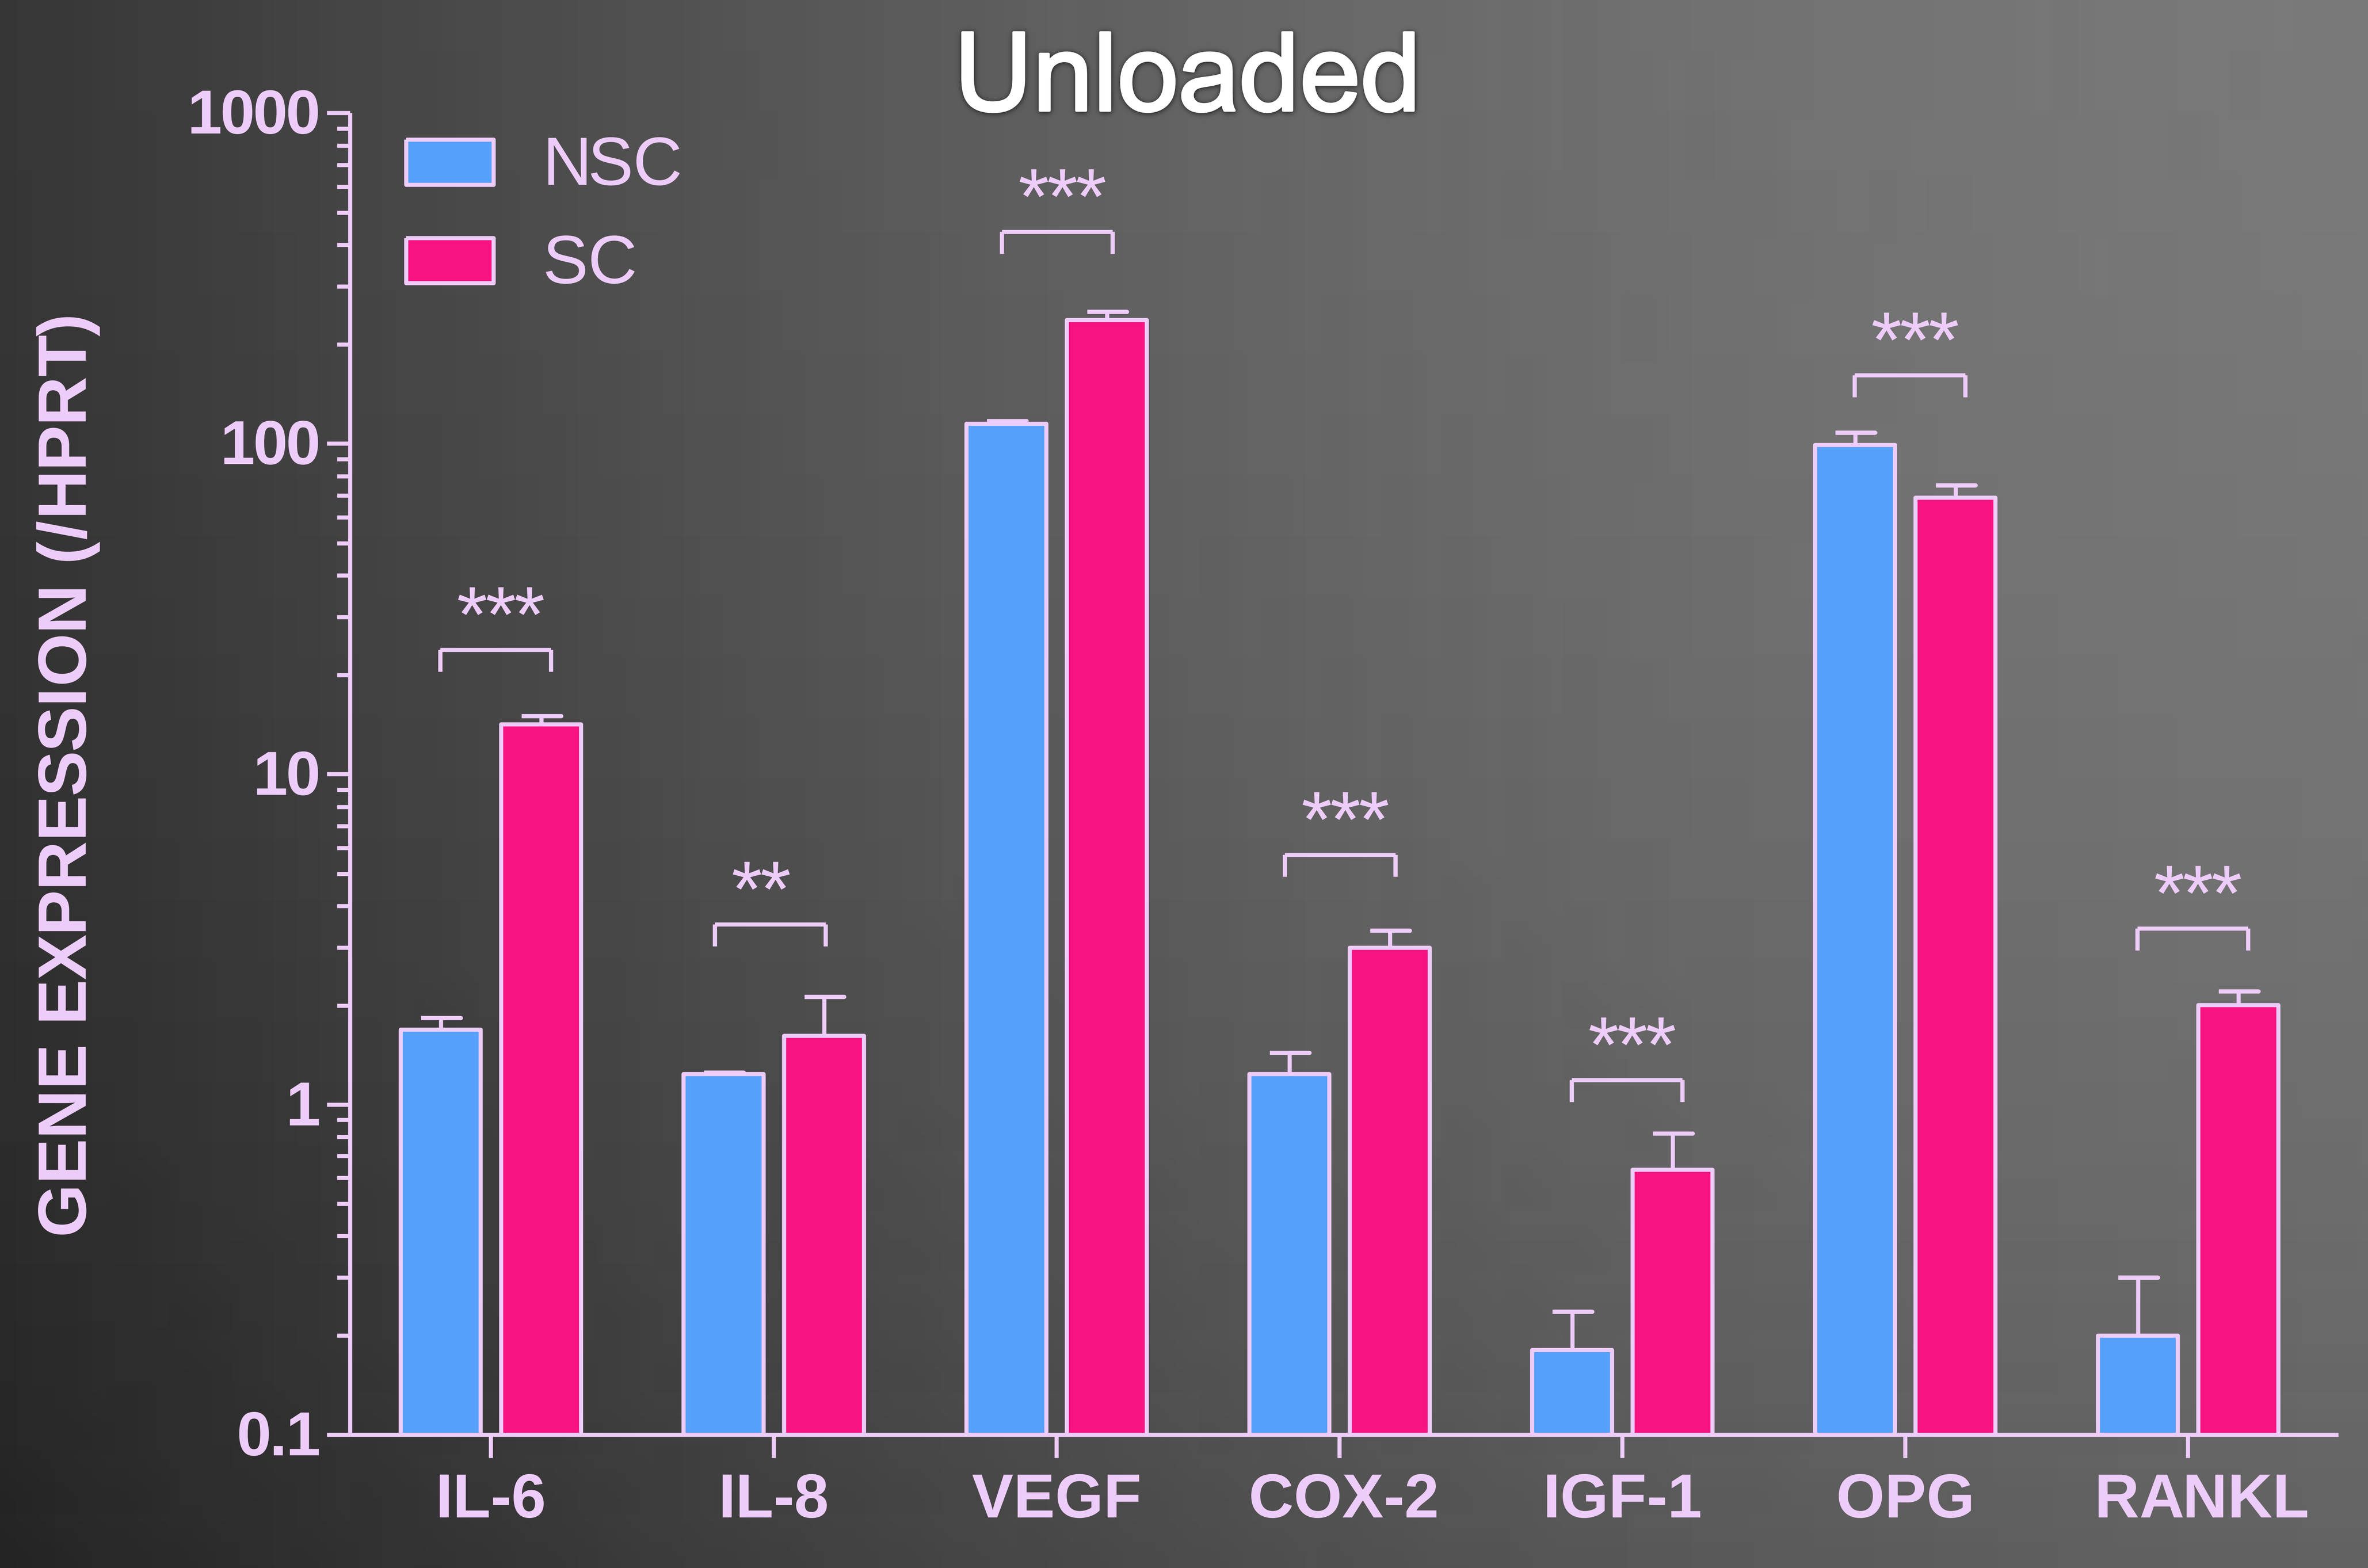
Unloaded
NSC
SC
GENE EXPRESSION (/HPRT)
1000
100
10
1
0.1

**

IL-6
IL-8
VEGF
COX-2
IGF-1
OPG
RANKL

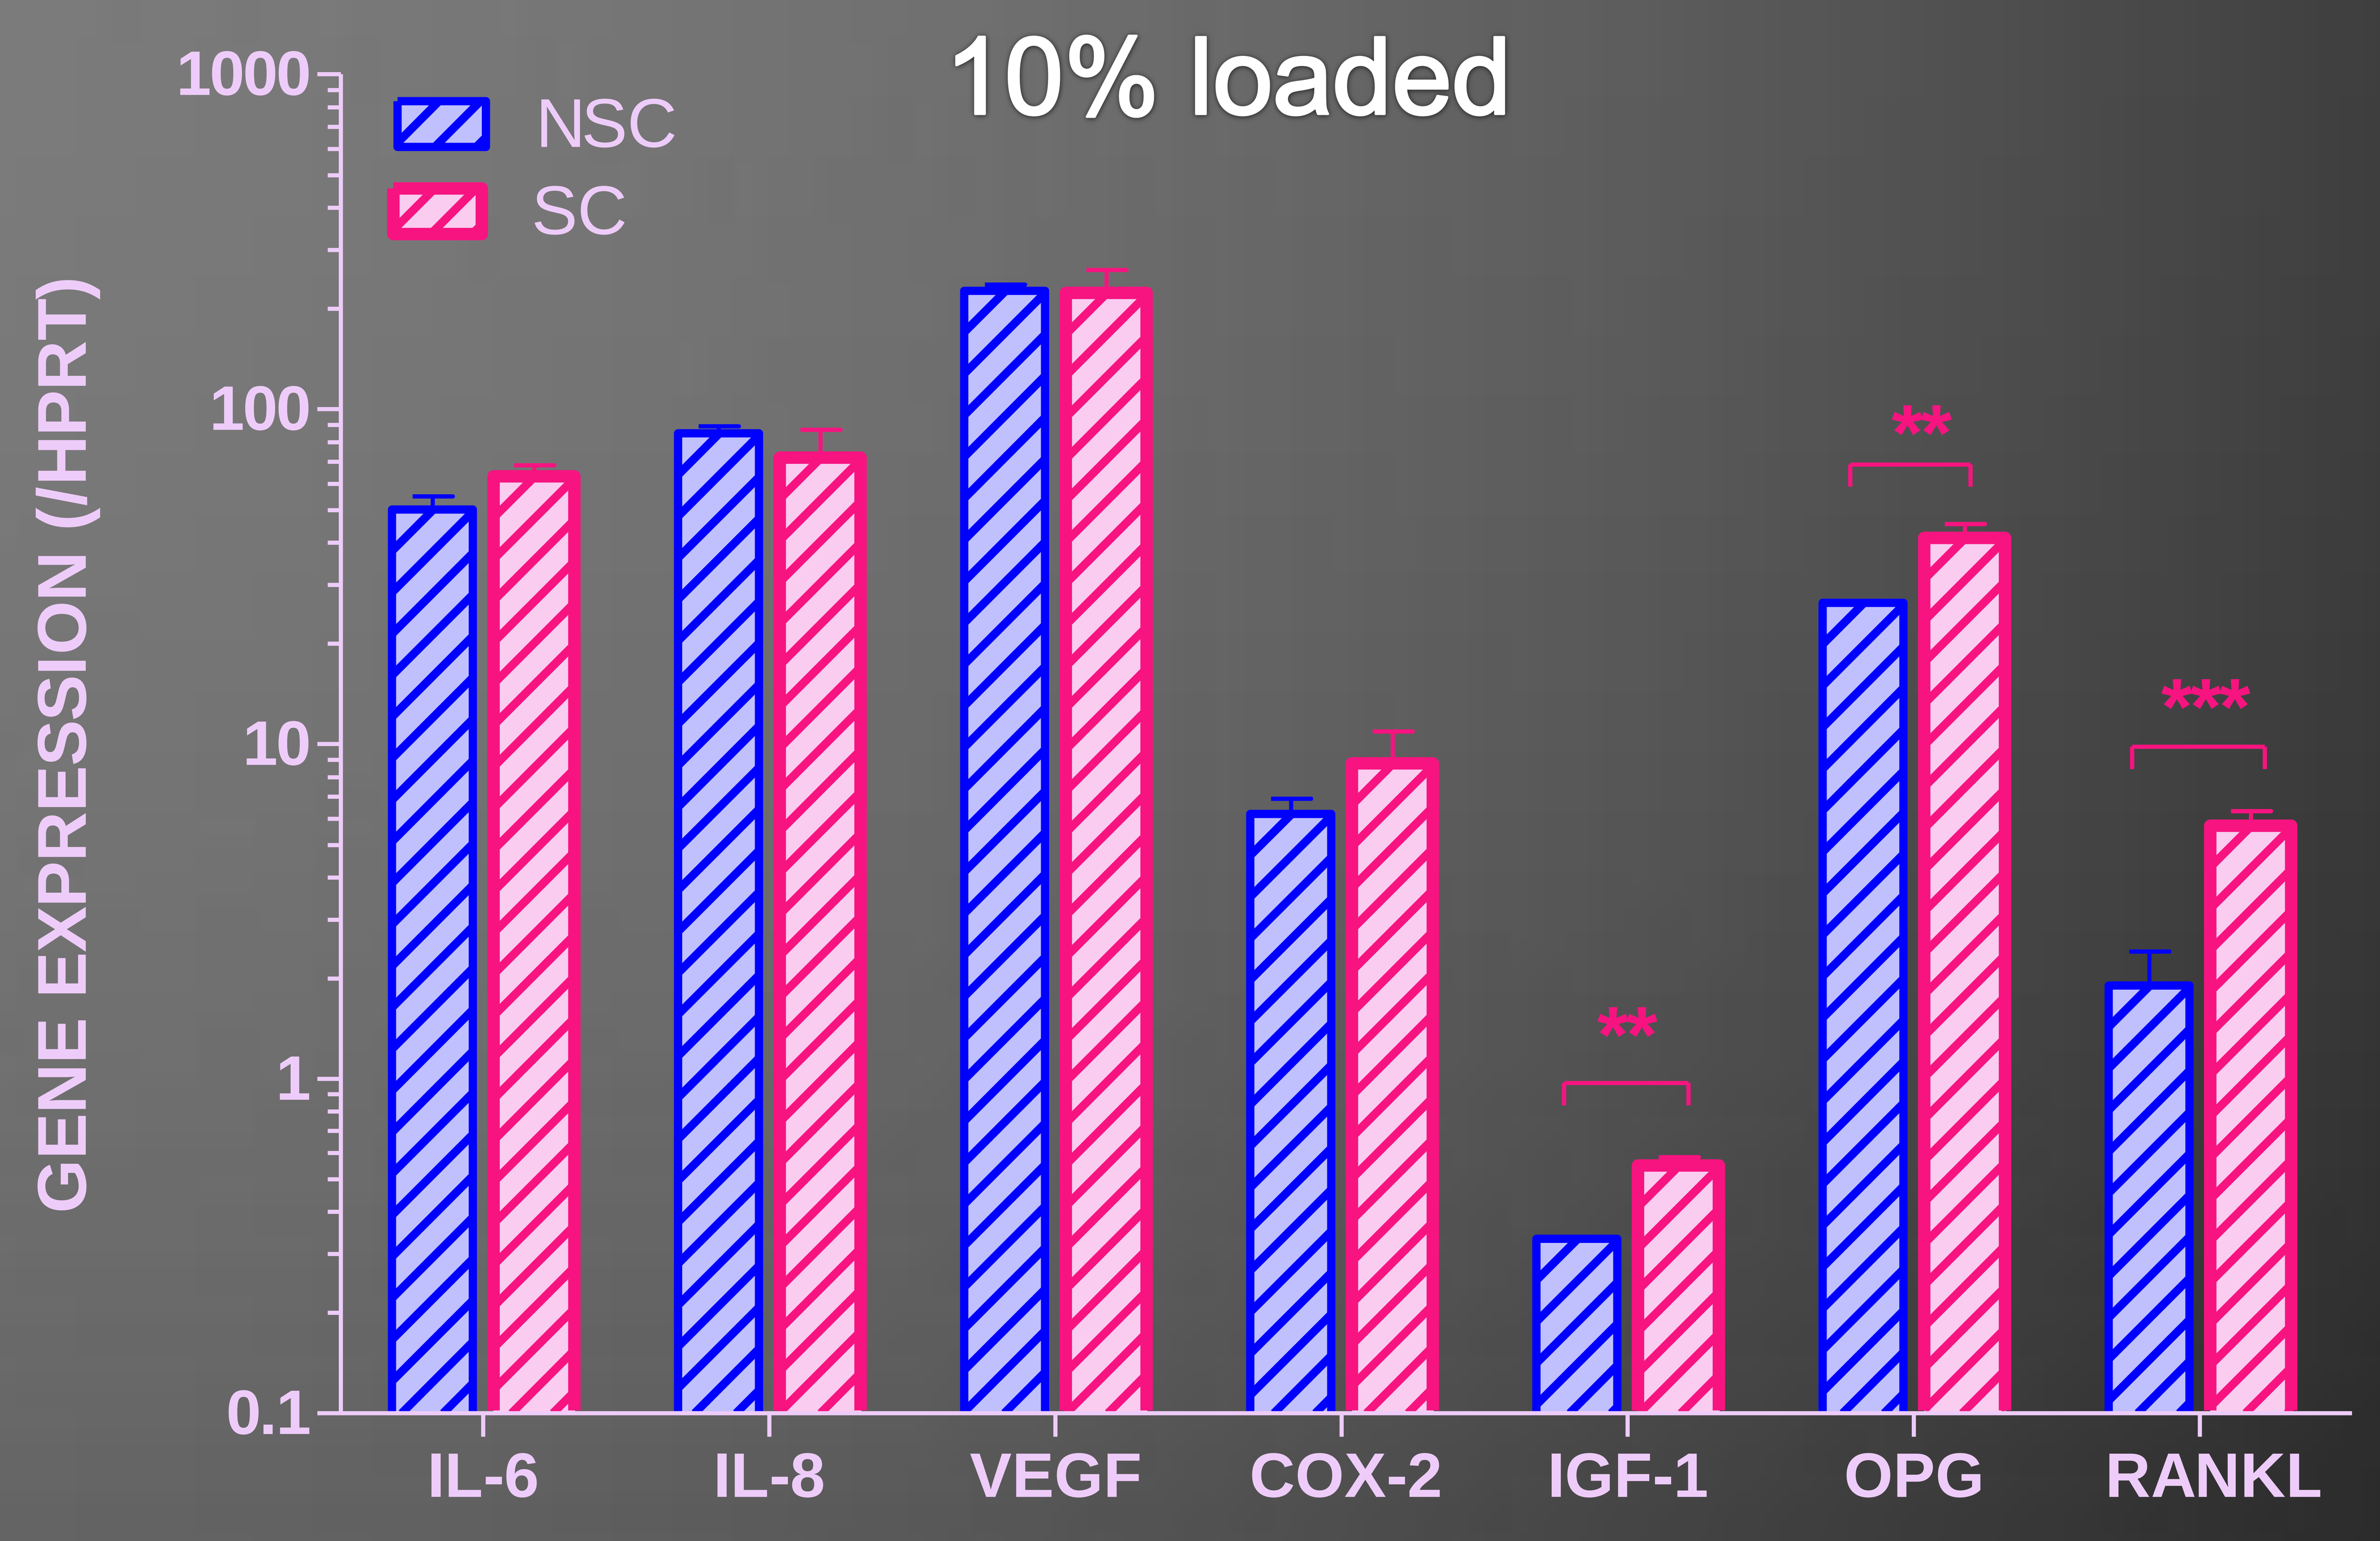
10% loaded
NSC
SC
GENE EXPRESSION (/HPRT)
1000
100
10
1
0.1
**
**

IL-6
IL-8
VEGF
COX-2
IGF-1
OPG
RANKL

Conclusions. These results indicate that in response to compression NSC osteoblasts expressed a phenotype similar to that of SC osteoblasts. Moreover, SC osteoblasts are less sensitive to mechanical stimuli than NSC osteoblasts. These results clarify the role of compression in the pathogenesis of subchondral bone sclerosis and allow new perspectives of research in this field.