Avocado/Soybean Unsaponifiables decrease the expression of pro-inflammatory genes in human chondrocytes

M Mathy-Hartert [1], P Devel [1], N Piccardi [2], J-Y Reginster [1], Henrotin Y [1]

[1] Bone and Cartilage Metabolism Research, University of Liege, Belgium.

[2] Laboratoires Expanscience™, Epernon, France.

AIM OF THE STUDY.

The progressive deterioration and loss of cartilage leading to joint impairment are the common final pathogenic events in arthritis. Cartilage degradation is induced by inflammatory mediators secreted by chondrocytes but also by cells of the inflamed synovium. The unsaponifiables part of avocado (A) and soybean (S) oils (ASU, Piascledine, Laboratories Expanscience, Courbevoie, France) has been used to treat OA for several years. They have been demonstrated to relieve symptoms in hip and knee OA and to prevent structural changes in hip OA. Our objective was to investigate the effects of ASU on the expression by human chondrocytes of pro-inflammatory genes including interleukin (IL) -6, -8, inducible nitric oxide synthase (iNOS) and cyclooxygenase 2 (COX2).

METHODS.

Human chondrocytes were enzymatically isolated from OA knee cartilage. Chondrocytes were cultured for 24 h in a well-defined culture medium in the presence or not of ASU mixed in a ratio 1:2 (A1S2, 1 part of avocado and 2 parts of soybean, 10 µg/ml). After washings, the cells were cultured for the next 24 h with or without IL-1beta (10^{-11} M). IL-6, IL-8, iNOS and COX-2 gene expressions were analysed by real time and quantitative RT PCR (LightCycler, Roche).

RESULTS.

In the basal conditions, OA chondrocytes spontaneously expressed low mRNA levels of IL-6, IL-8, iNOS and COX-2. IL-1beta increased IL-6, IL-8, iNOS, COX-2 mRNA levels by 41, 210, 6.3 and 6.2 times respectively. Pre-incubation with A1S2 significantly down-regulated IL-1beta-induced IL-6 ($p < 0.01$), IL-8 ($p < 0.001$), iNOS ($p < 0.001$) and COX-2 ($p < 0.001$) gene expression. The basal expression of iNOS and COX2 was also decreased ($p<0,001$).

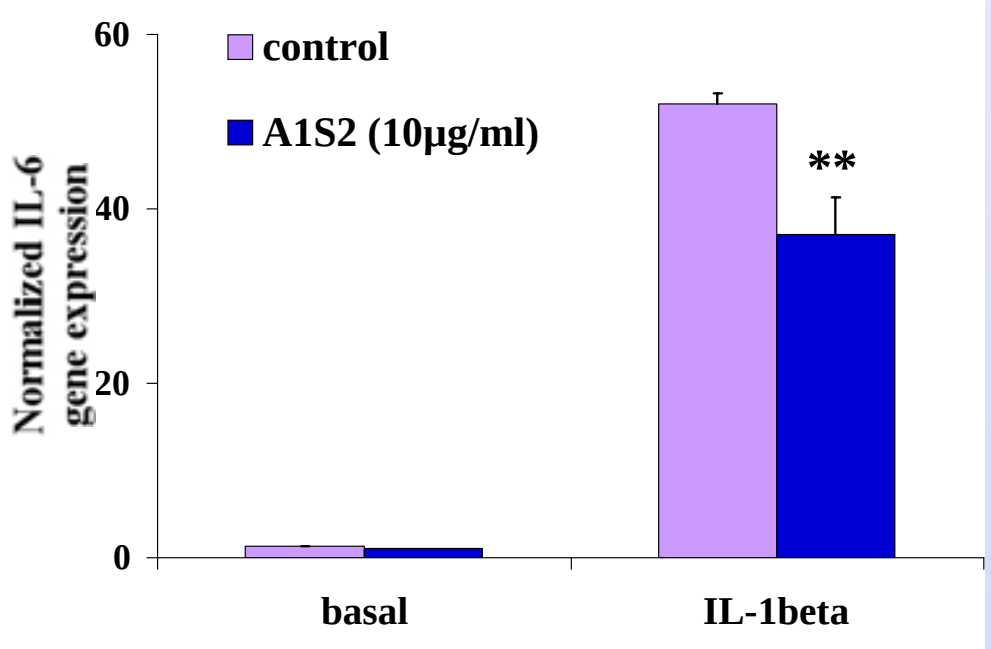
60
40
20
0
control
A1S2 (10µg/ml)
**
Normalized IL-6 gene expression
basal
IL-1beta

Effect of A1S2 on IL-6 gene expression

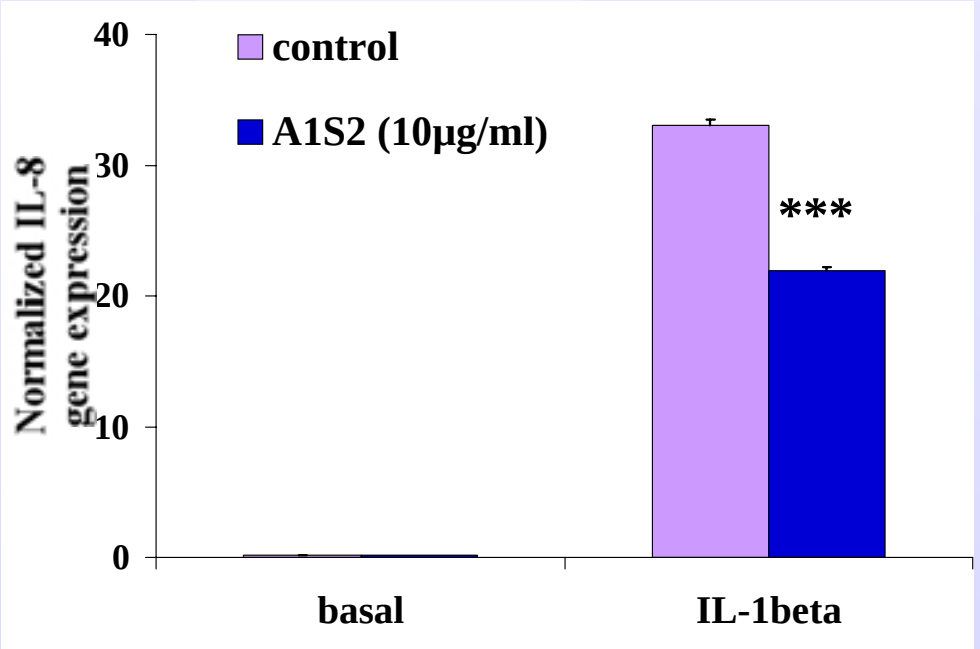
40
30
20
10
0
control
A1S2 (10µg/ml)

Normalized IL-8 gene expression
basal
IL-1beta

Effect of A1S2 on IL-8 gene expression

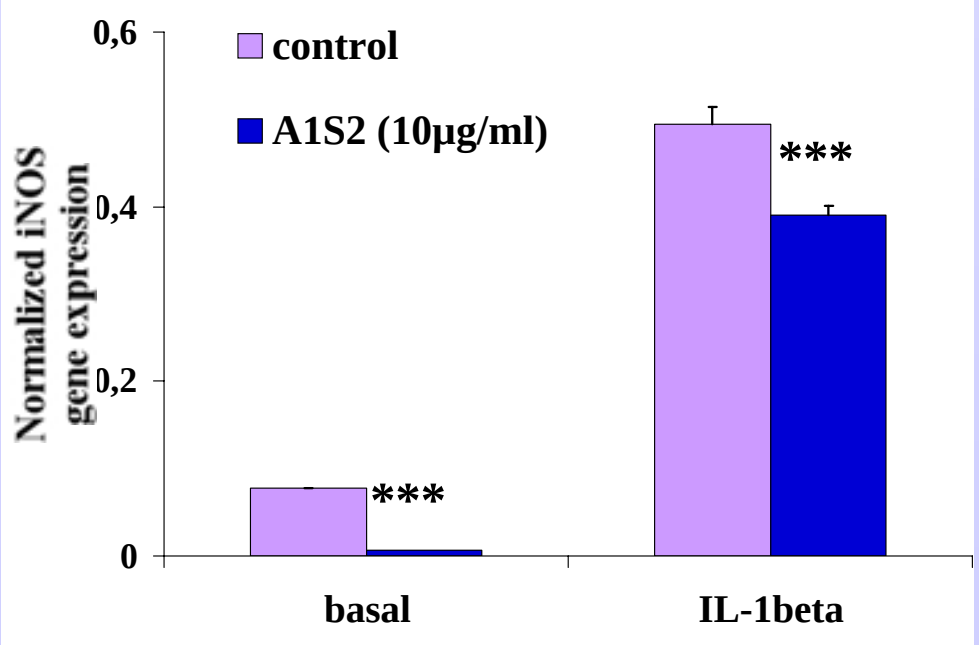
0,6
0,4
0,2
0
control
A1S2 (10µg/ml)

Normalized iNOS gene expression
basal
IL-1beta

Effect of A1S2 on iNOS gene expression

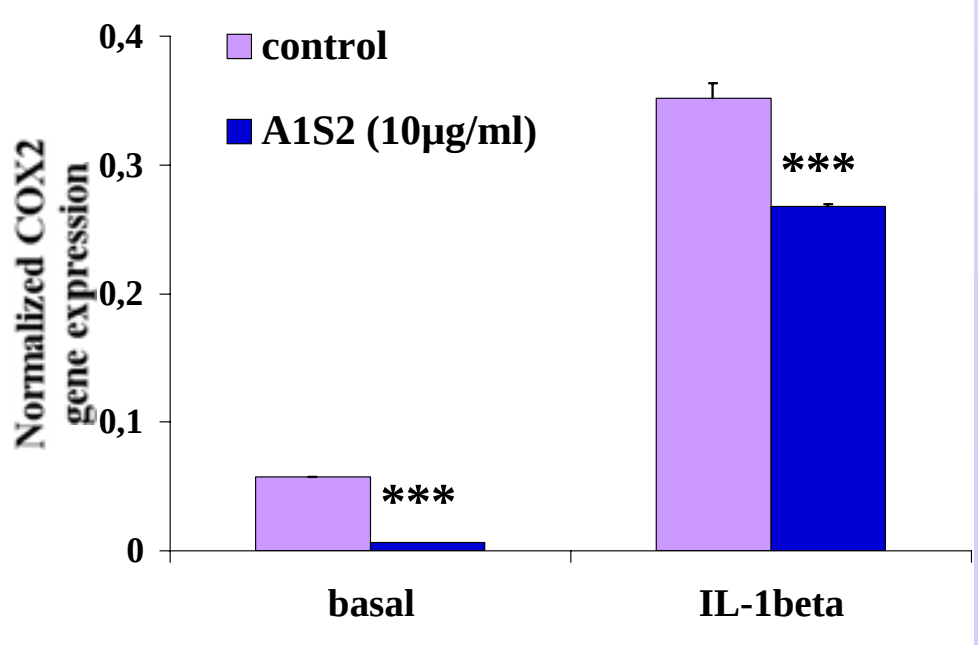
0,4
0,3
0,2
0,1
0
control
A1S2 (10µg/ml)

Normalized COX2 gene expression
basal
IL-1beta

Effect of A1S2 on COX2 gene expression

CONCLUSIONS.

These data demonstrate that ASU may have anti-inflammatory properties by counteracting the stimulating effects of IL-1beta on pro-inflammatory genes. These findings contribute to explain the symptomatic effects observed in OA patients.